SAFETY AND IMMUNOGENICITY OF TYPHOID FEVER AND YELLOW FEVER VACCINES WHEN ADMINISTERED CONCOMITANTLY WITH QUADRIVALENT MENINGOCOCCAL ACWY GLYCOCONJUGATE VACCINE IN HEALTHY ADULTS

Martin Alberer MD[1], Gerd Burchard MD[2], Tomas Jelinek MD[3], Emil Reisinger MD[4], Jiri Beran MD[5], Lucie Hlavata MD[6], Eduardo Forleo-Neto MD[7], Alemnew F Dagnew MD[7], Ashwani K Arora MD[8]

[1] Department of Infectious Diseases and Tropical Medicine, University of Munich, Munich, Germany; [2] University Medical Center Hamburg-Eppendorf, Hamburg, Germany; [3] Berlin Center for Travel and Tropical Medicine, Berlin, Germany; [4] Department of Tropical Medicine and Infectious Diseases, University of Rostock Medical School, Rostock, Germany; [5] Vaccination and Travel Medicine Centre, Hradec Kralove, Czech Republic; [6] Novartis Vaccines, Prague, Czech Republic; [7] Novartis Vaccines, Cambridge, USA; [8] Novartis Vaccines, Sienna, Italy

BACKGROUND & OBJECTIVES

- It is often desirable to complete pre-travel immunization in a single visit and simultaneous administration of vaccines is recommended.[1] Providing vaccines concomitantly will simplify the process of acquiring pre-travel prophylaxis and may help ensure that all required vaccines are administered.
- However, concomitant vaccination could potentially compromise immunogenicity and/or safety of the individual vaccines and, therefore, possible vaccine interaction should be carefully assessed.
- This phase 3b randomized, open-label study assessed the immunogenicity and safety of travel vaccines including vaccines for typhoid fever (TF) and yellow fever (YF) when administered with or without a quadrivalent meningococcal glycoconjugate vaccine (MenACWY-CRM) (NCT01466387). Results of concomitant administration of MenACWY-CRM with Japanese encephalitis and rabies vaccination are reported separately.

METHODS

- In this study, overall 552 healthy adults aged 18 through 60 years were enrolled, of which 301 subjects were randomized to 1 of 3 vaccine regimens presented here.
 All vaccines were administered on day 1.
 - TF + YF + MenACWY-CRM (n=100)
 - TF + YF (n=101)
 - MenACWY-CRM (n=100).
- Immunogenicity at baseline and 4 weeks (day 29) following vaccination was assessed by serum bactericidal assay using human complement (hSBA), enzyme-linked immunosorbent assay (ELISA) or neutralization test.
 - The primary immunogenicity objective was to demonstrate non-inferiority of the immune responses to TF + YF + MenACWY-CRM regimen as compared to TF + YF alone, as measured by geometric mean titers/concentrations (GMTs/GMCs) to TF and YF at day 29 post-vaccination.
 - Secondary immunogenicity objectives were to evaluate the seroprotection rates elicited by YF vaccine at day 29 post-vaccination and to assess the immune responses to MenACWY-CRM vaccine given concomitantly with TF and YF vaccines versus given alone.
- Adverse events (AEs), serious AEs (SAEs) and AEs leading to study withdrawal were recorded throughout the study period.

RESULTS

- The three vaccine groups were comparable with respect to age, weight and height. Subjects were predominantly Caucasian and 50% to 56% across groups were males (**Table 1**).
- Of all subjects, 99% to 100% across groups completed the study protocol. Reasons for premature withdrawal were loss to follow-up (n=1) and protocol deviation (n=1).

Table 1: Baseline demographics and characteristics

	TF+YF+MenACWY-CRM n=100	TF+YF n=101	MenACWY-CRM n=100
Age, years (Mean±SD)	35.0±11.0	36.5±10.8	36.9±11.2
Male, n (%)	56 (56%)	52 (51%)	50 (50%)
Female, n (%)	44 (44%)	49 (49%)	50 (50%)
Ethnicity, n (%)			
Asian, n (%)	1 (1%)	1 (<1%)	0
Black or African American, n (%)	3 (3%)	0	0
Caucasian, n (%)	96 (96%)	99 (98%)	99 (99%)
Other, n (%)	0	1 (<1%)	1 (1)
Weight, kg (Mean±SD)	77.7±18.1	77.8±15.7	76.1±15.9
Height, cm (Mean±SD)	175.2±9.5	173.6±10.2	173.8±9.7
Met entry criteria, n (%)	100 (100%)	100 (99%)	100 (100%)

SD, standard deviation.

IMMUNOGENICITY

- Pre-specified criteria for non-inferiority of post-vaccination GMCs and GMTs were met for TF and YF vaccines, respectively, when given concomitantly with MenACWY-CRM vaccine versus when given alone. At day 29 post-vaccination, the lower limits of the 95% confidence interval for GMT/GMC group ratios were above the non-inferiority margin of 0.5: TF GMC group ratio: 1.14 (95% CI: **0.81**-1.6), YF GMT group ratio: 0.96 (95% CI **0.65**-1.41) (**Figure 1**).
- Post-vaccination TF GMCs were 153 (TF + YF + MenACWY-CRM) and 134 (TF + YF). YF GMTs following vaccination were 5022 (TF + YF + MenACWY-CRM) and 5244 (TF + YF) (**Figure 1**).
- The percentages of subjects with seroprotective anti-YF neutralizing antibody titers ≥1/10 at day 29 post-vaccination were 97% when TF and YF vaccines were administered with MenACWY-CRM and 100% when these vaccines were administered without MenACWY-CRM vaccine (**Figure 2**).
- Percentages of subjects achieving post-vaccination hSBA titers ≥8 for meningococcal serogroups A, C, W-135 and Y were 77%, 76%, 97%, and 91% (TF + YF + MenACWY-CRM) and 76%, 77%, 95%, and 89% (MenACWY-CRM group), respectively (**Figure 3A**).
- Post-vaccination GMTs against serogroups A, C, W-135 and Y were generally in the same range when MenACWY-CRM vaccine was given alone or co-administered with TF and YF vaccines (**Figure 3B**).

Figure 1. GMTs against YF, GMC against TF (days 1 and 29) and day 29 post-vaccination ratio of GMCs/GMTs (95% CI) for TF and YF travel vaccines given simultaneously with MenACWY-CRM vaccine versus given alone. Criteria for non-inferiority: lower limit of 95% confidence interval should be >0.5

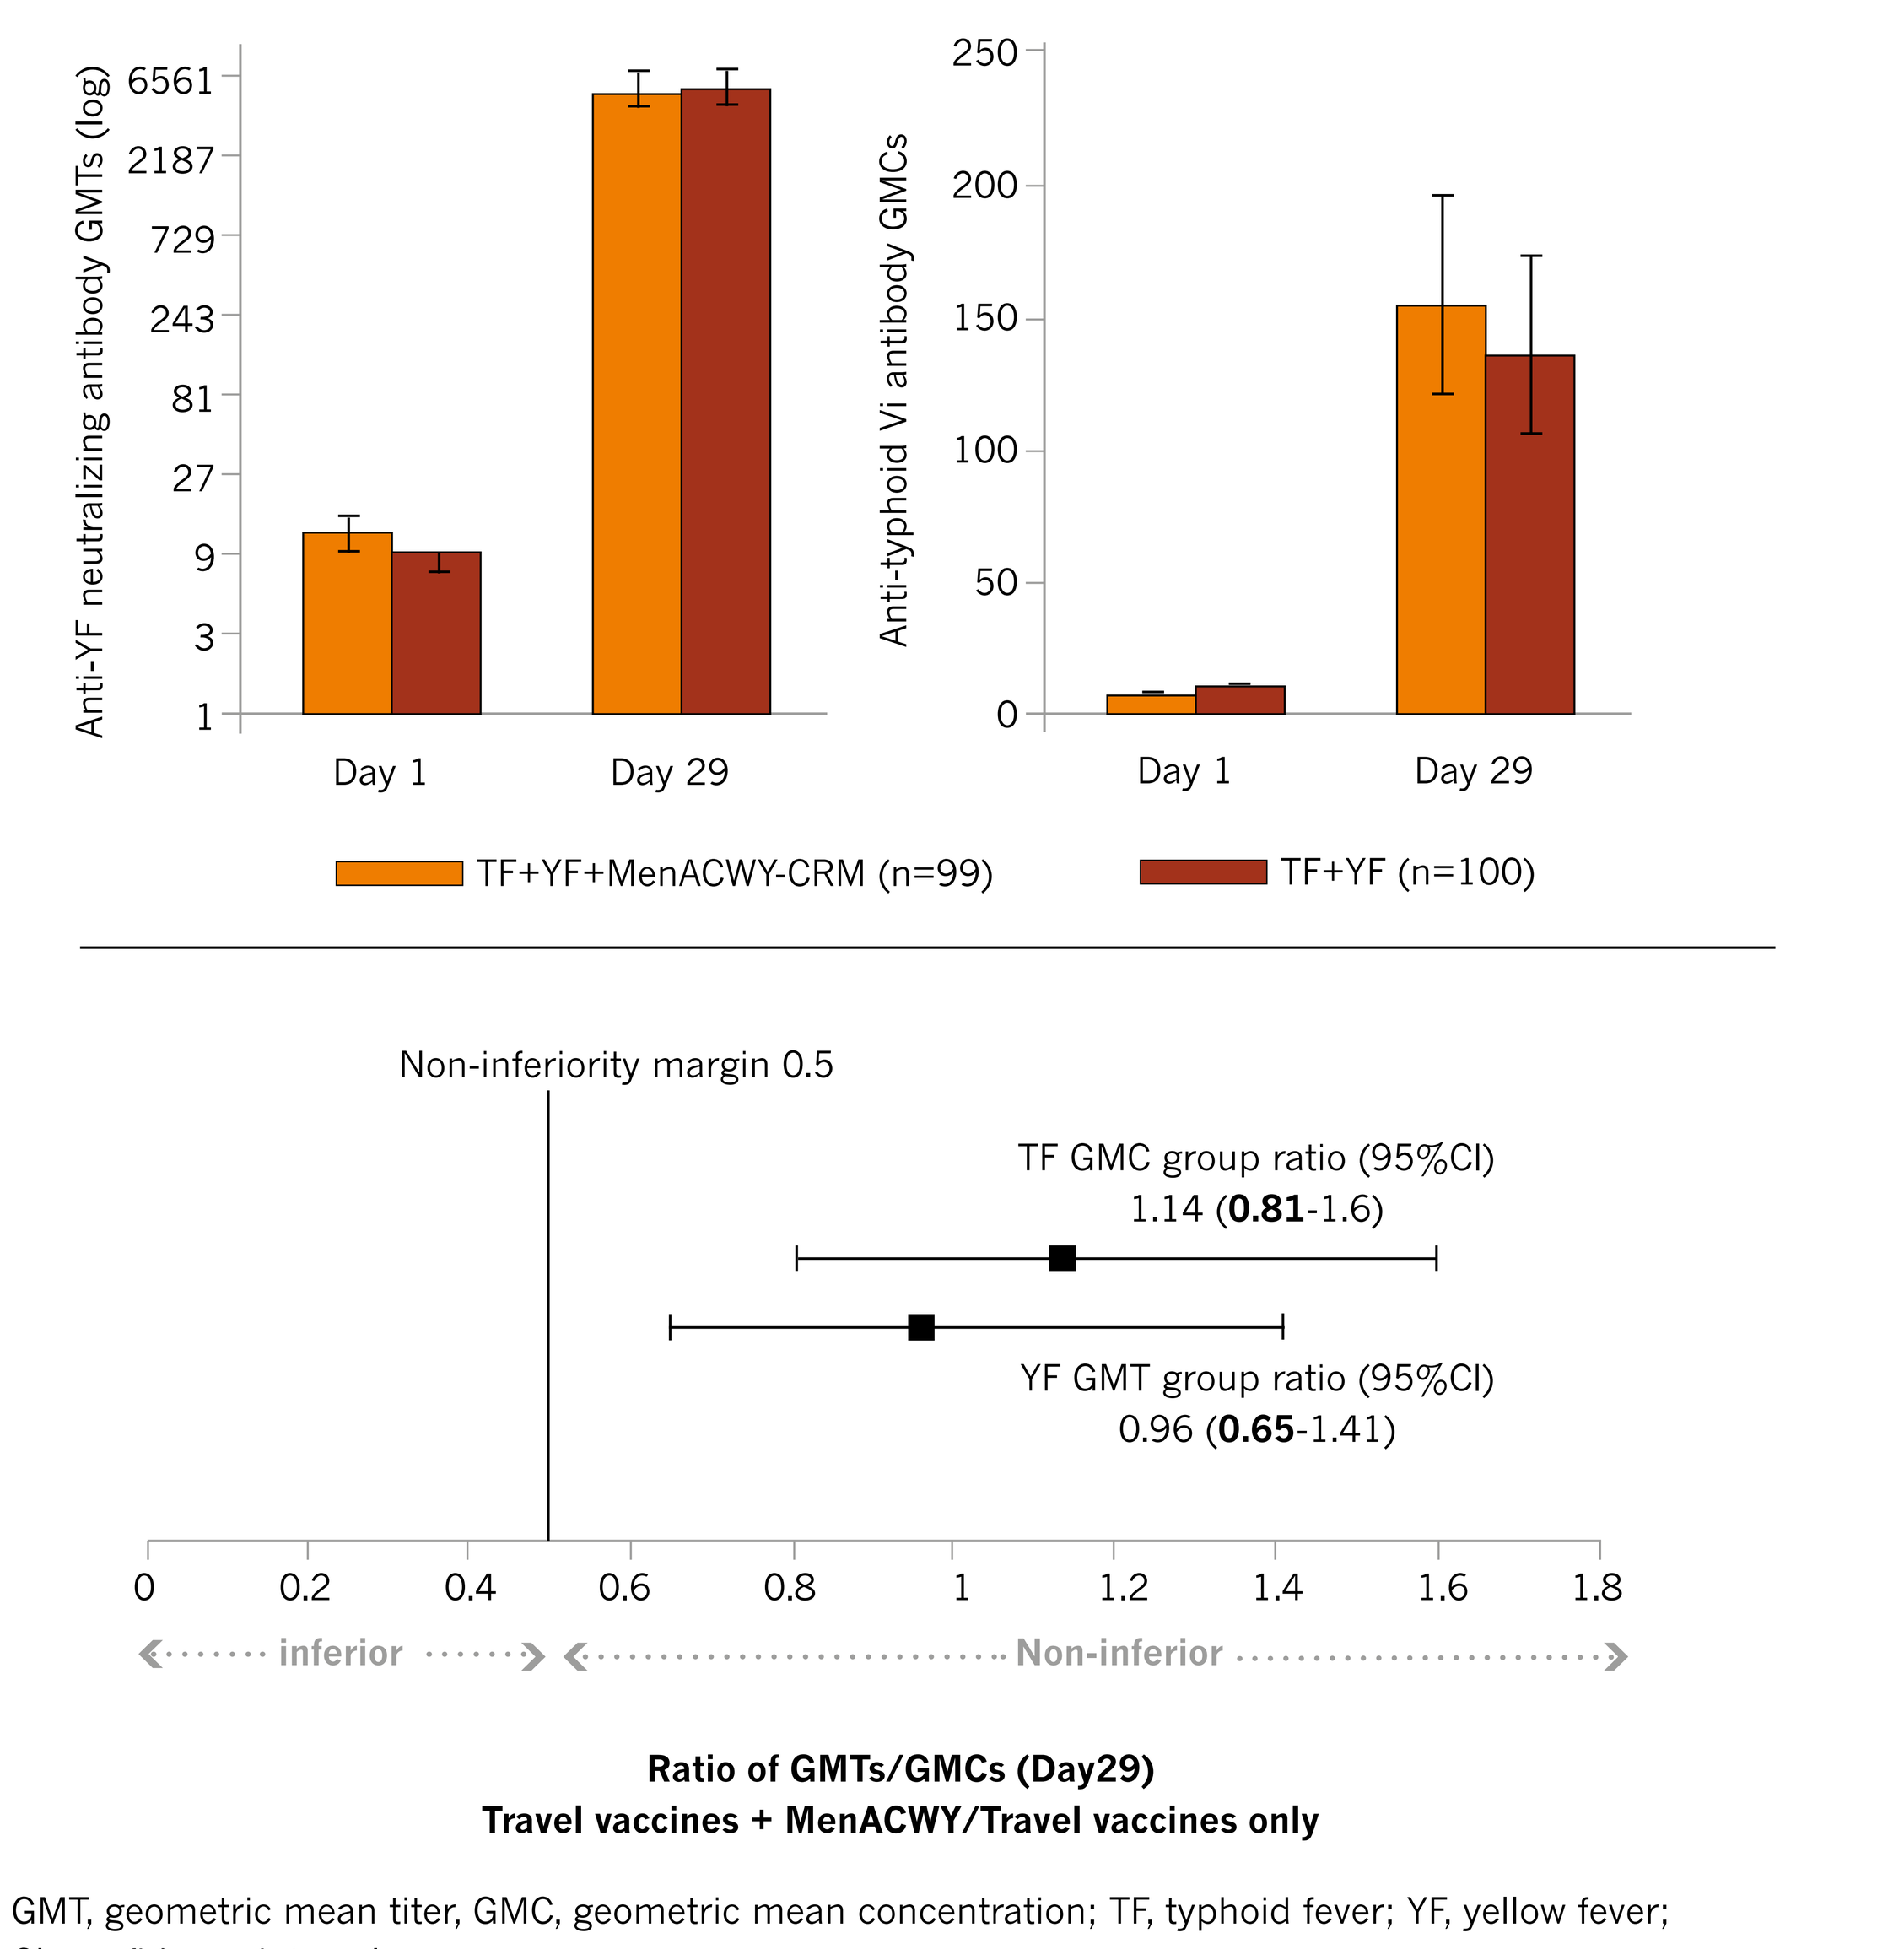
Anti-YF neutralizing antibody GMTs (log)
6561
2187
729
243
81
27
9
3
1
Day 1
Day 29
Anti-typhoid Vi antibody GMCs
250
200
150
100
50
0
Day 1
Day 29
TF+YF+MenACWY-CRM (n=99)
TF+YF (n=100)
Non-inferiority margin 0.5
TF GMC group ratio (95%CI)
1.14 (0.81-1.6)
YF GMT group ratio (95%CI)
0.96 (0.65-1.41)
0
0.2
0.4
0.6
0.8
1
1.2
1.4
1.6
1.8
inferior
Non-inferior
Ratio of GMTs/GMCs (Day29)
Travel vaccines + MenACWY/Travel vaccines only

GMT, geometric mean titer, GMC, geometric mean concentration; TF, typhoid fever; YF, yellow fever; CI, confidence interval

Figure 2. Percentage of subjects (95% CI) achieving anti-yellow fever neutralizing antibody titers ≥1/10 at baseline and day 29 post-vaccination, by vaccine regimen

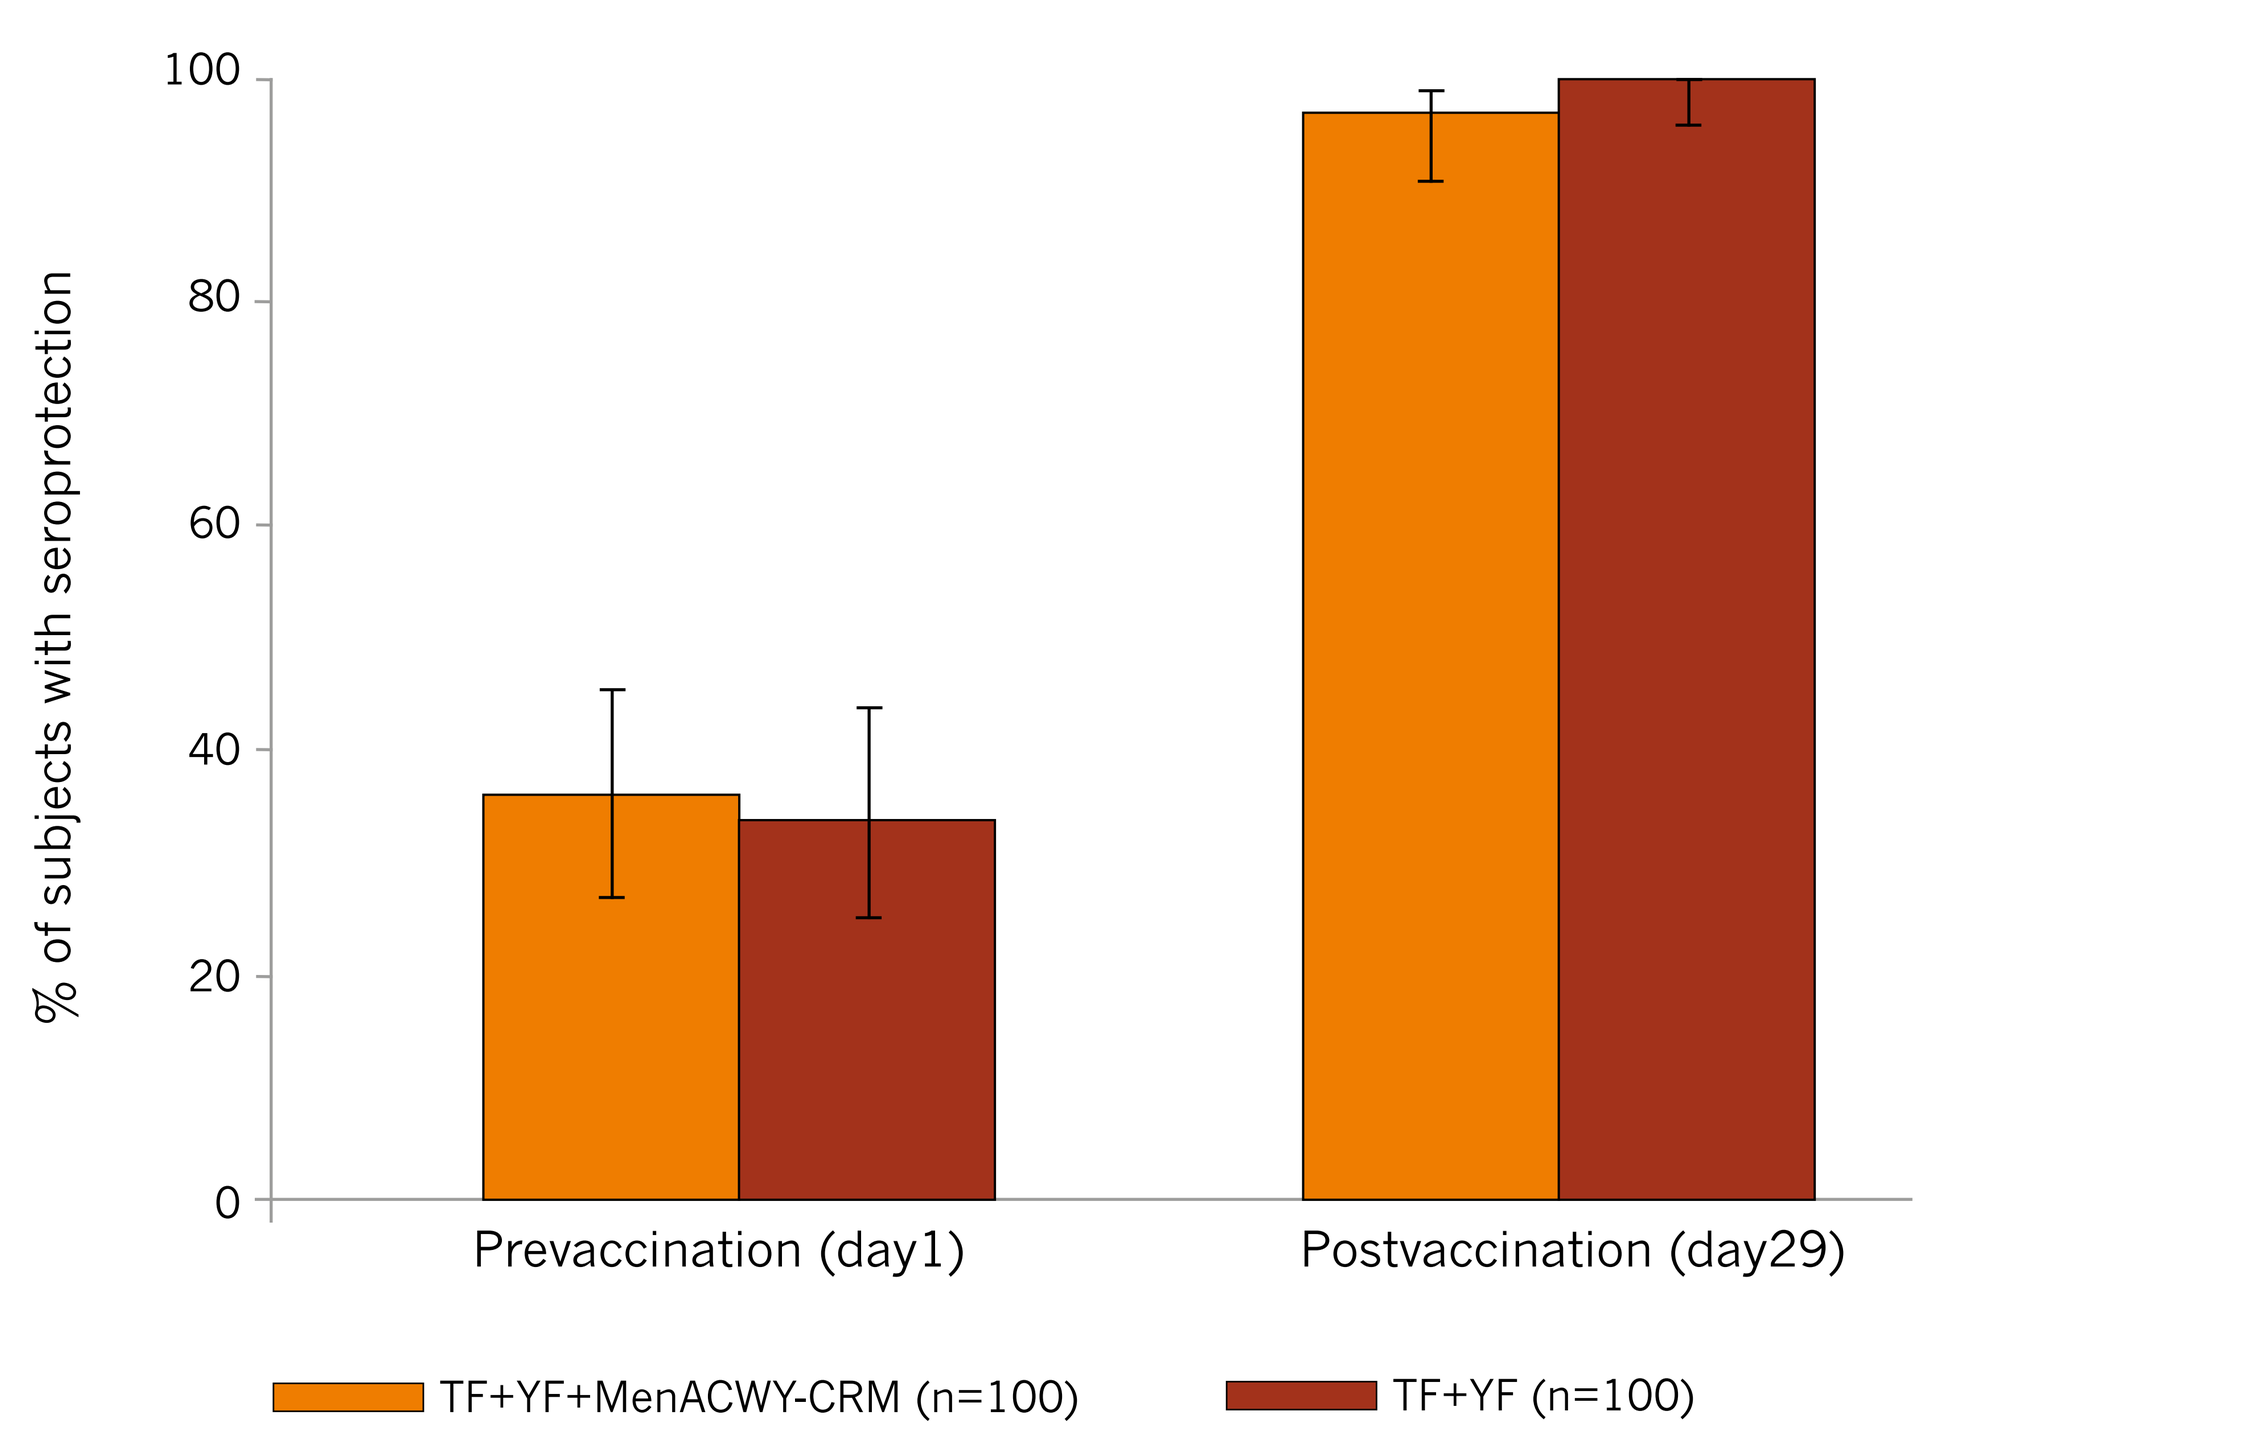
% of subjects with seroprotection
100
80
60
40
20
0
Prevaccination (day1)
Postvaccination (day29)
TF+YF+MenACWY-CRM (n=100)
TF+YF (n=100)

TF, typhoid fever; YF yellow fever; CI, confidence interval

Figure 3. Percentages of subjects with hSBA titers ≥8 at day 29 post-vaccination (A) and GMTs at baseline and day 29 post-vaccination (B) (95% CI) against meningococcal serogroups A, C, W-135 and Y, by vaccine regimen

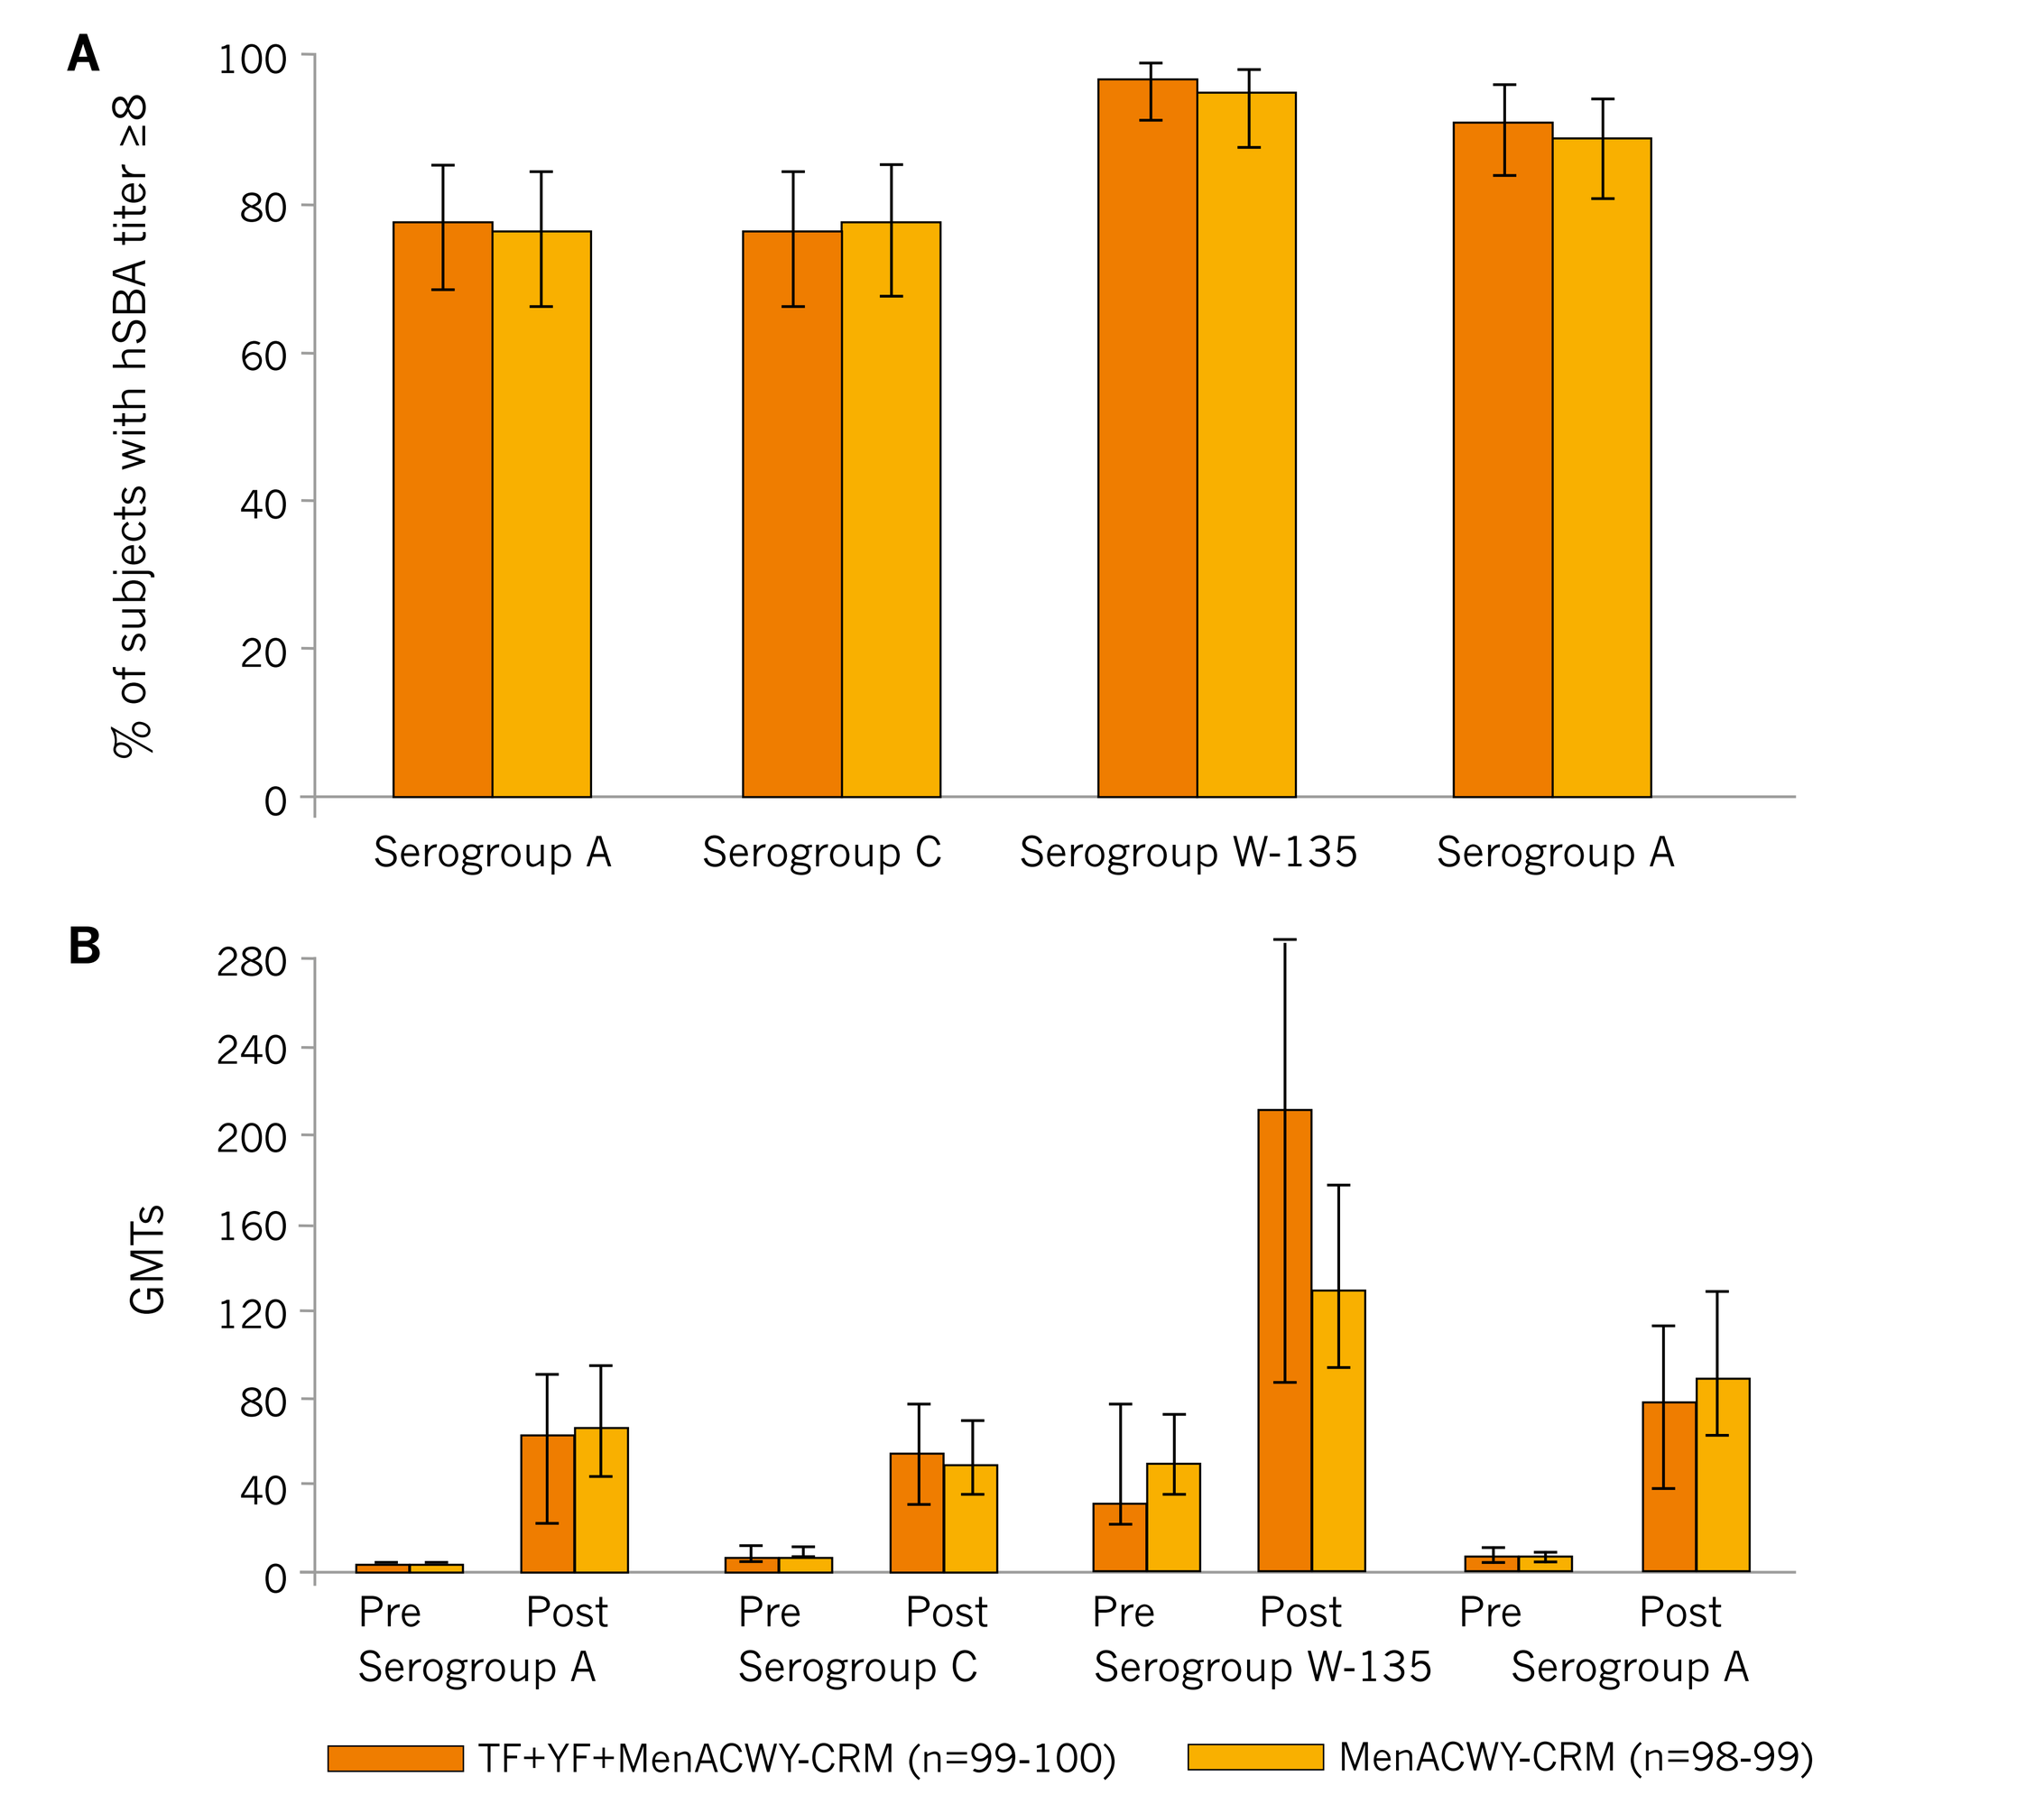
A
% of subjects with hSBA titer ≥8
100
80
60
40
20
0
Serogroup A
Serogroup C
Serogroup W-135
Serogroup A
B
GMTs
280
240
200
160
120
80
40
0
Pre
Post
Serogroup A
Pre
Post
Serogroup C
Pre
Post
Serogroup W-135
Pre
Post
Serogroup A
TF+YF+MenACWY-CRM (n=99-100)
MenACWY-CRM (n=98-99)

GMT, geometric mean titer; TF, typhoid fever; YF yellow fever; CI, confidence interval

SAFETY

- The percentage of subjects reporting AEs was the same for TF and YF vaccines with or without MenACWY-CRM vaccine.
- Any AEs were observed in 41% of subjects in groups TF + YF + MenACWY-CRM and TF + YF; 30% to 34% of these events were considered at least possibly related to study vaccination. In the MenACWY group, 14% of subjects reported any AE, of which 8% were considered at least possibly related (**Table 2**).
- By preferred term, the most common at least possibly related AEs in TF + YF + MenACWY-CRM and TF + YF groups were injection site pain (8% and 12%) and headache (10% and 7%), respectively. In MenACWY-CRM, the most common possibly related AEs were headache (4%) and injection site pain (2%).
- Most of the AEs were of mild to moderate severity, with only 2 to 3 subjects across groups reporting AEs of severe intensity.
- There were no SAEs, premature study withdrawals or deaths in any of the vaccination regimens (**Table 2**).

Table 2: Number and percentage of subjects with spontaneous adverse events (AEs)

	TF + YF + MenACWY-CRM	TF + YF	MenACWY-CRM
n (%)	**n=100**	**n=101**	**n=100**
Any AEs	41 (41%)	41 (41%)	14 (14%)
At least possibly related AEs	34 (34%)	30 (30%)	8 (8%)
SAEs	0	0	0
At least possibly related SAEs	0	0	0
Any AE leading to premature withdrawal	0	0	0
Deaths	0	0	0

TF, typhoid fever; YF, yellow fever; AE, adverse event; SAE, serious adverse event.

CONCLUSION

- MenACWY-CRM can be administered concomitantly with typhoid Vi polysaccharide vaccine and live attenuated yellow fever vaccine without compromising antibody responses or safety to these individual vaccines and can be incorporated into existing pre-travel vaccination programmes.

REFERENCE

1 General recommendations on immunization. Recommendations of the Advisory Committee on Immunization Practices (ACIP). Morbidity and mortality weekly report Recommendations and reports (MWRR) / Centers for Disease Control 2011; 60(2): 1-64

NOVARTIS VACCINES

5th Northern European Conference on Travel Medicine
June 5th – 8th 2014 Bergen, Norway

Contact information:
ashwani_kumar.arora@novartis.com